Effects of SM on myelination, axonal density, oligodendrogenesis and astrogenesis in EAE and on BMP signaling pathways

Maya Golan[1], Moshe Ben Hamou[2], Karin Fainberg[1], Nadav Bleich Kimelman[3], Shai Rubnov[3], Uri Danon[3], Ehud Marom3, Arnon Karni* [1,2,4]
[1]Tel Aviv Sourasky Medical Center, Neurology, Tel Aviv, Israel, [2]Faculty of Medicine, Tel Aviv University, Tel Aviv, Israel, [3]Stem Cell Medicine , Jerusalem, Israel, [4]Tel Aviv University, Sagol School of Neuroscience, Tel Aviv , Israel

BACKGROUND

Multiple sclerosis (MS) pathology is characterized by remyelination failure despite the presence of oligodendrocyte progenitor cells (OPCs), leaving axons permanently demyelinated and vulnerable to degeneration. Despite the presence of OPCs in demyelinated MS lesions, their differentiation into functional oligodendrocytes is insufficient, and most lesions evolve into nonfunctional astroglial scars. We have previously reported that parenteral anti bone morphogenetic protein (BMP) blocking therapy ameliorated demyelinating diseases in animal such as experimental autoimmune encephalomyelitis (EAE) and cuprizone fed mice through oligodendrogenesis and remyelination. We have reported that novel anti-BMP small molecule (SM) improves the clinical severity of EAE, induced oligodendrogenesis and decreased phospho-SMAD expression in the spinal cord (SC) tissue, given both i.p. and oraly. Here we have further characterized the effect of the SM therapy on myelination axonal integrity, oligodendrogenesis and astrogenesis and studied its effect on BMPs signaling.

OBJECTIVE

To evaluate the effect of SM on the myelination, axonal integrity, oligodendrogenesis and astrogenesis and on the canonical and non-canonical signaling of BMPs.

METHODS

A novel anti-BMP- SM (750 mg/day), was given daily by oral gavage from day 9 (signs onset) until day 38 to an EAE induced SJL mice by PLP$_{139-151}$ vs. vehicle (PEG-400). The affected SC of these EAE mice were immunohistochemically stained to evaluating the differentiation of oligodendrocytes, astrocytes, myelination, and axon density by staining to CC1$^+$Olig2$^+$BrdU$^+$, GFAP$^+$Olig2$^+$BrdU$^+$, fluoromyelin (FM), and neurofilament (NF), respectively. Digital images were collected using confocal microscopy (Zeiss 710), and the number of double-positive cells was determined in 6-8 sections from each mouse (3 mice per group). The number of double-positive cells was quantified using *ImageJ* software and calculated as the number of cells/mm^2. Single-stained cells are presented as the percentage of the stained area.

We used Western blot analysis to examine the effects of SM on the canonical (phospho-SMAD 1,5,9) and non-canonical (phospho-p38 and phosphor-ERK 1,2) signaling pathways of BMP2, BMP4, BMP5 and BMP7 in the NIH3T3 cell line.

RESULTS

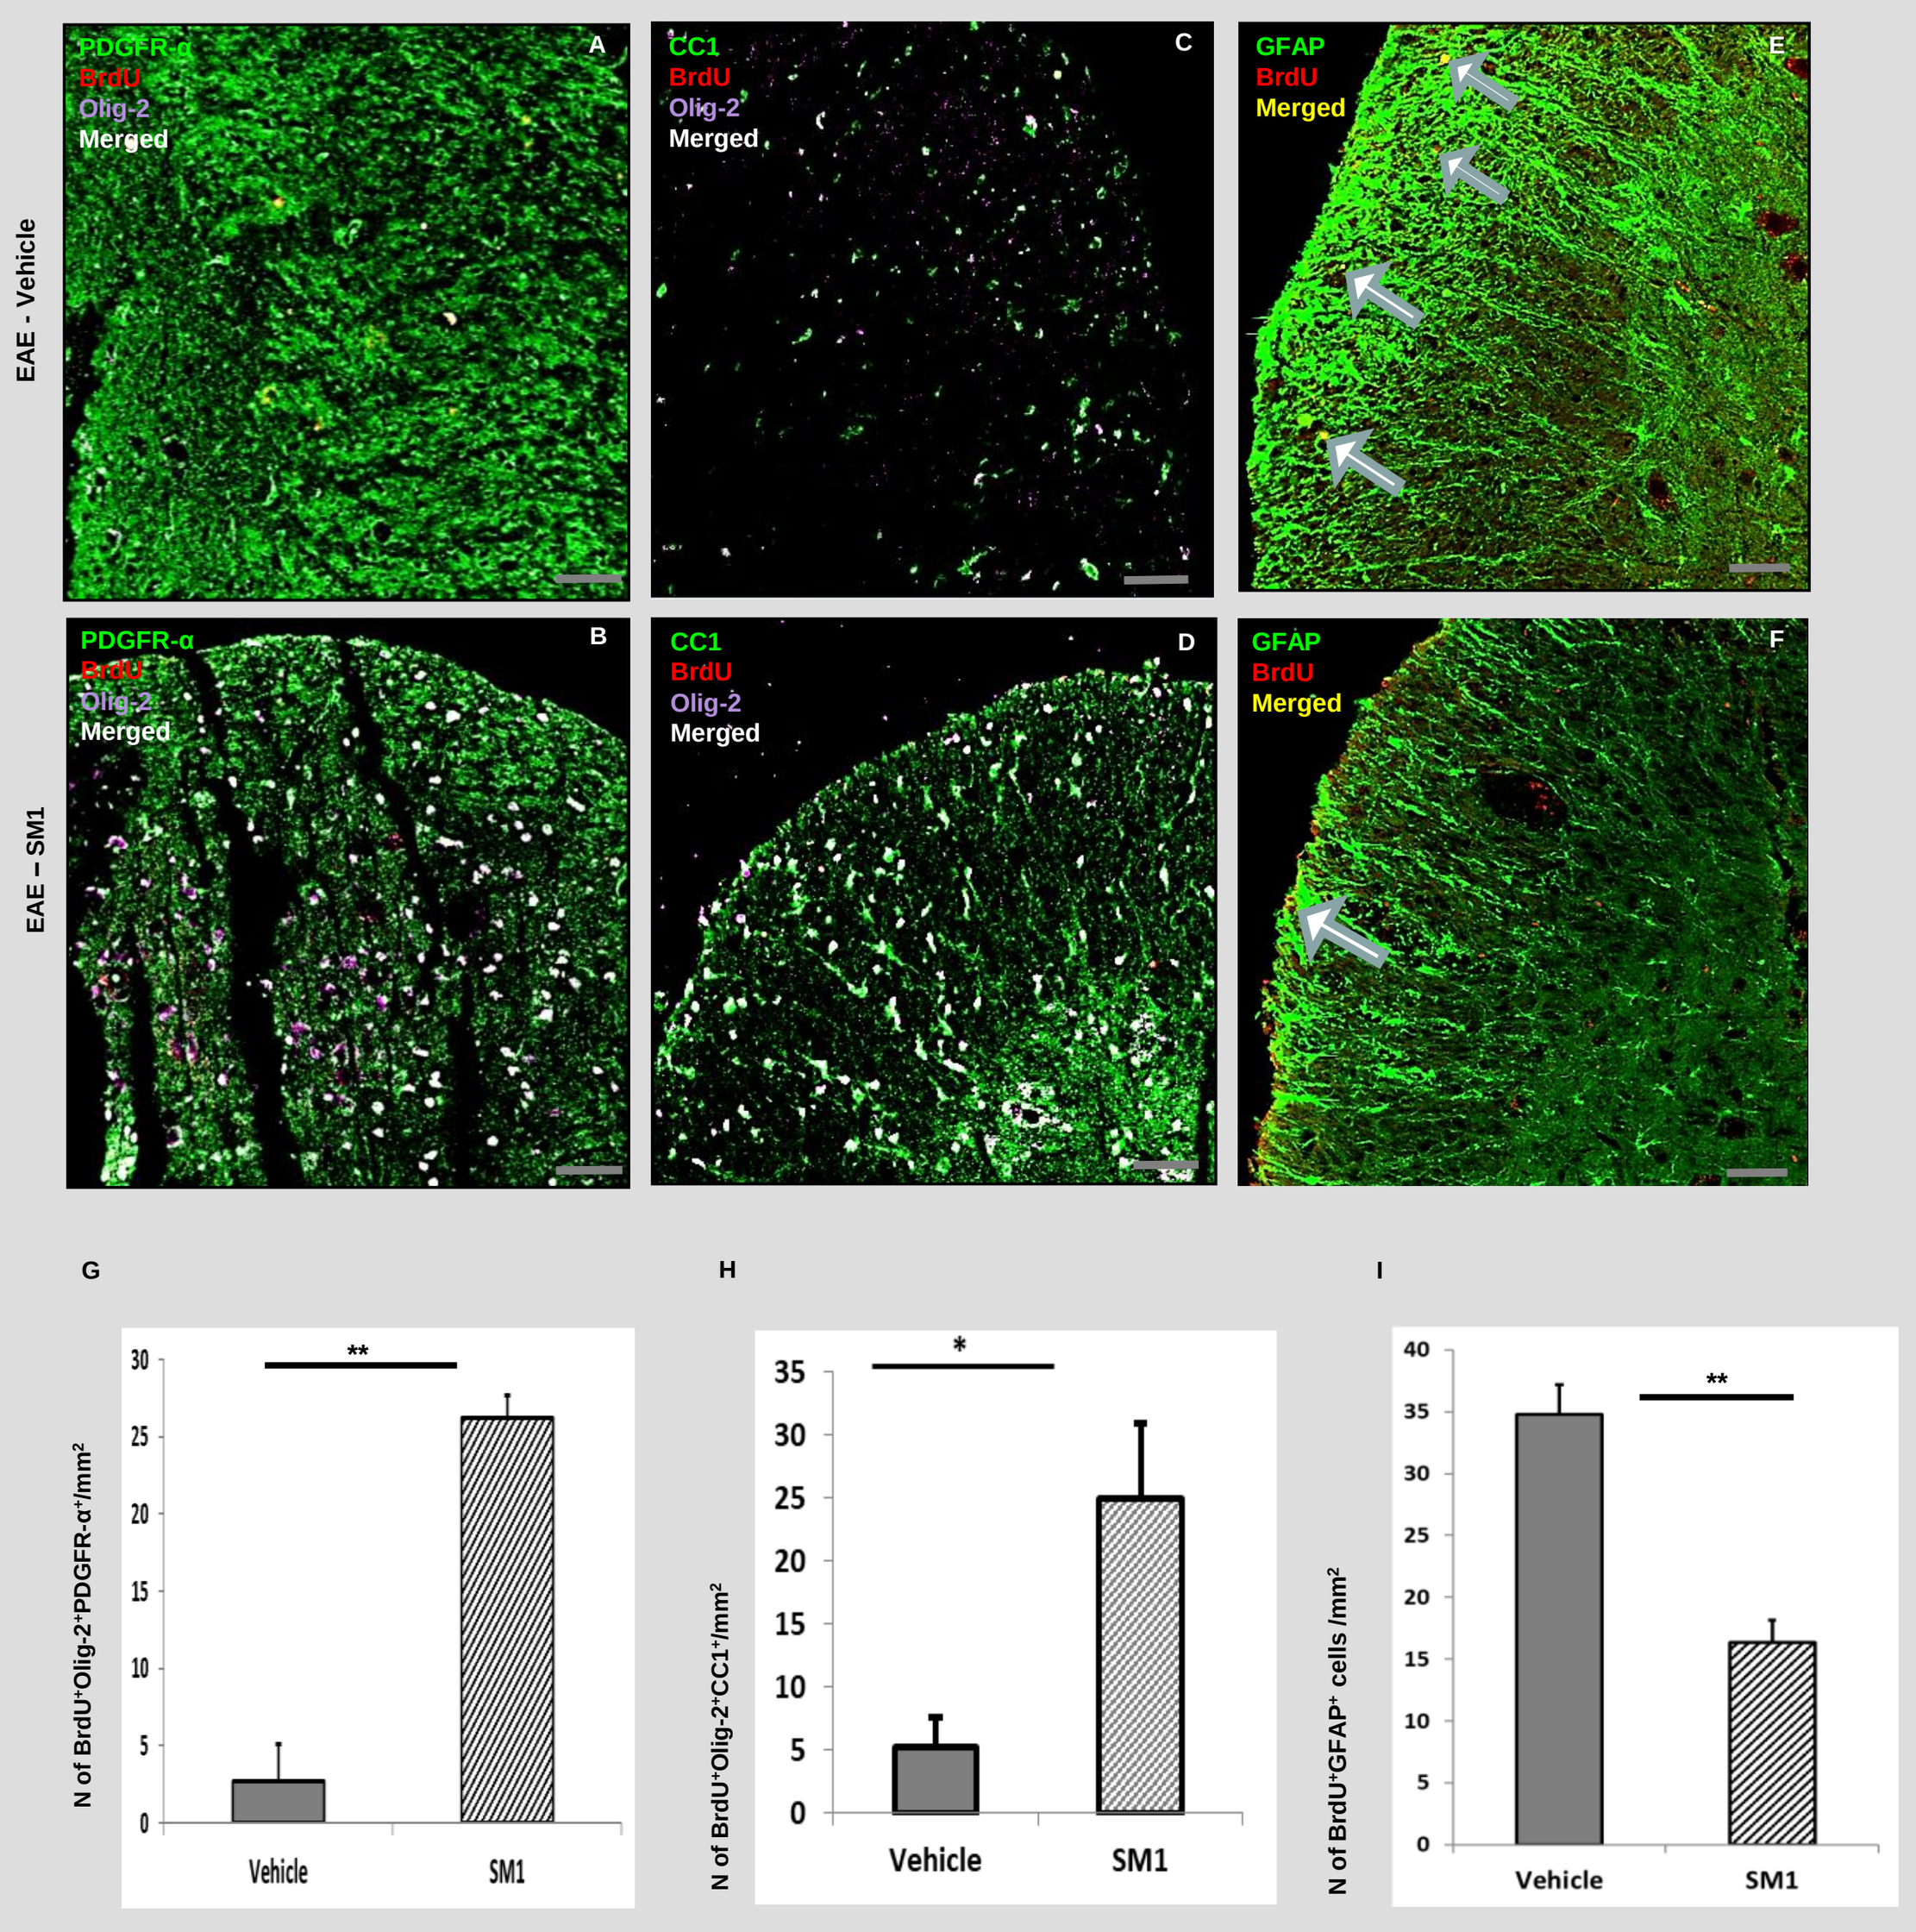
EAE - Vehicle
EAE – SM1
PDGFR-α
BrdU
Olig-2
Merged
CC1
GFAP
N of BrdU⁺Olig-2⁺PDGFR-α⁺/mm²
N of BrdU⁺Olig-2⁺CC1⁺/mm²
N of BrdU⁺GFAP⁺ cells /mm²
Vehicle
SM1

Figure 1. ***Daily oral administration of SM to EAE mice induces newly forms oligodendrocytes and reduces new astrocytes.*** Oral daily treatment with SM significantly increases the number of (A, B) premature (PDGFR-a$^+$Olig2$^+$BrdU$^+$) and (C,D) mature (CC1$^+$ Olig2$^+$BrdU$^+$) oligodendrocytes (OL) and (E,F) decreases the number of (GFAP$^+$BrdU$^+$) astrocytes compared to vehicle treated mice. Number of double positive cells/mm^2 is analyzed for premature OL (G), mature OL (H) and astrocytes (I). Scale bars= 50µm; * $p < 0.05$, ** $p < 0.01$.

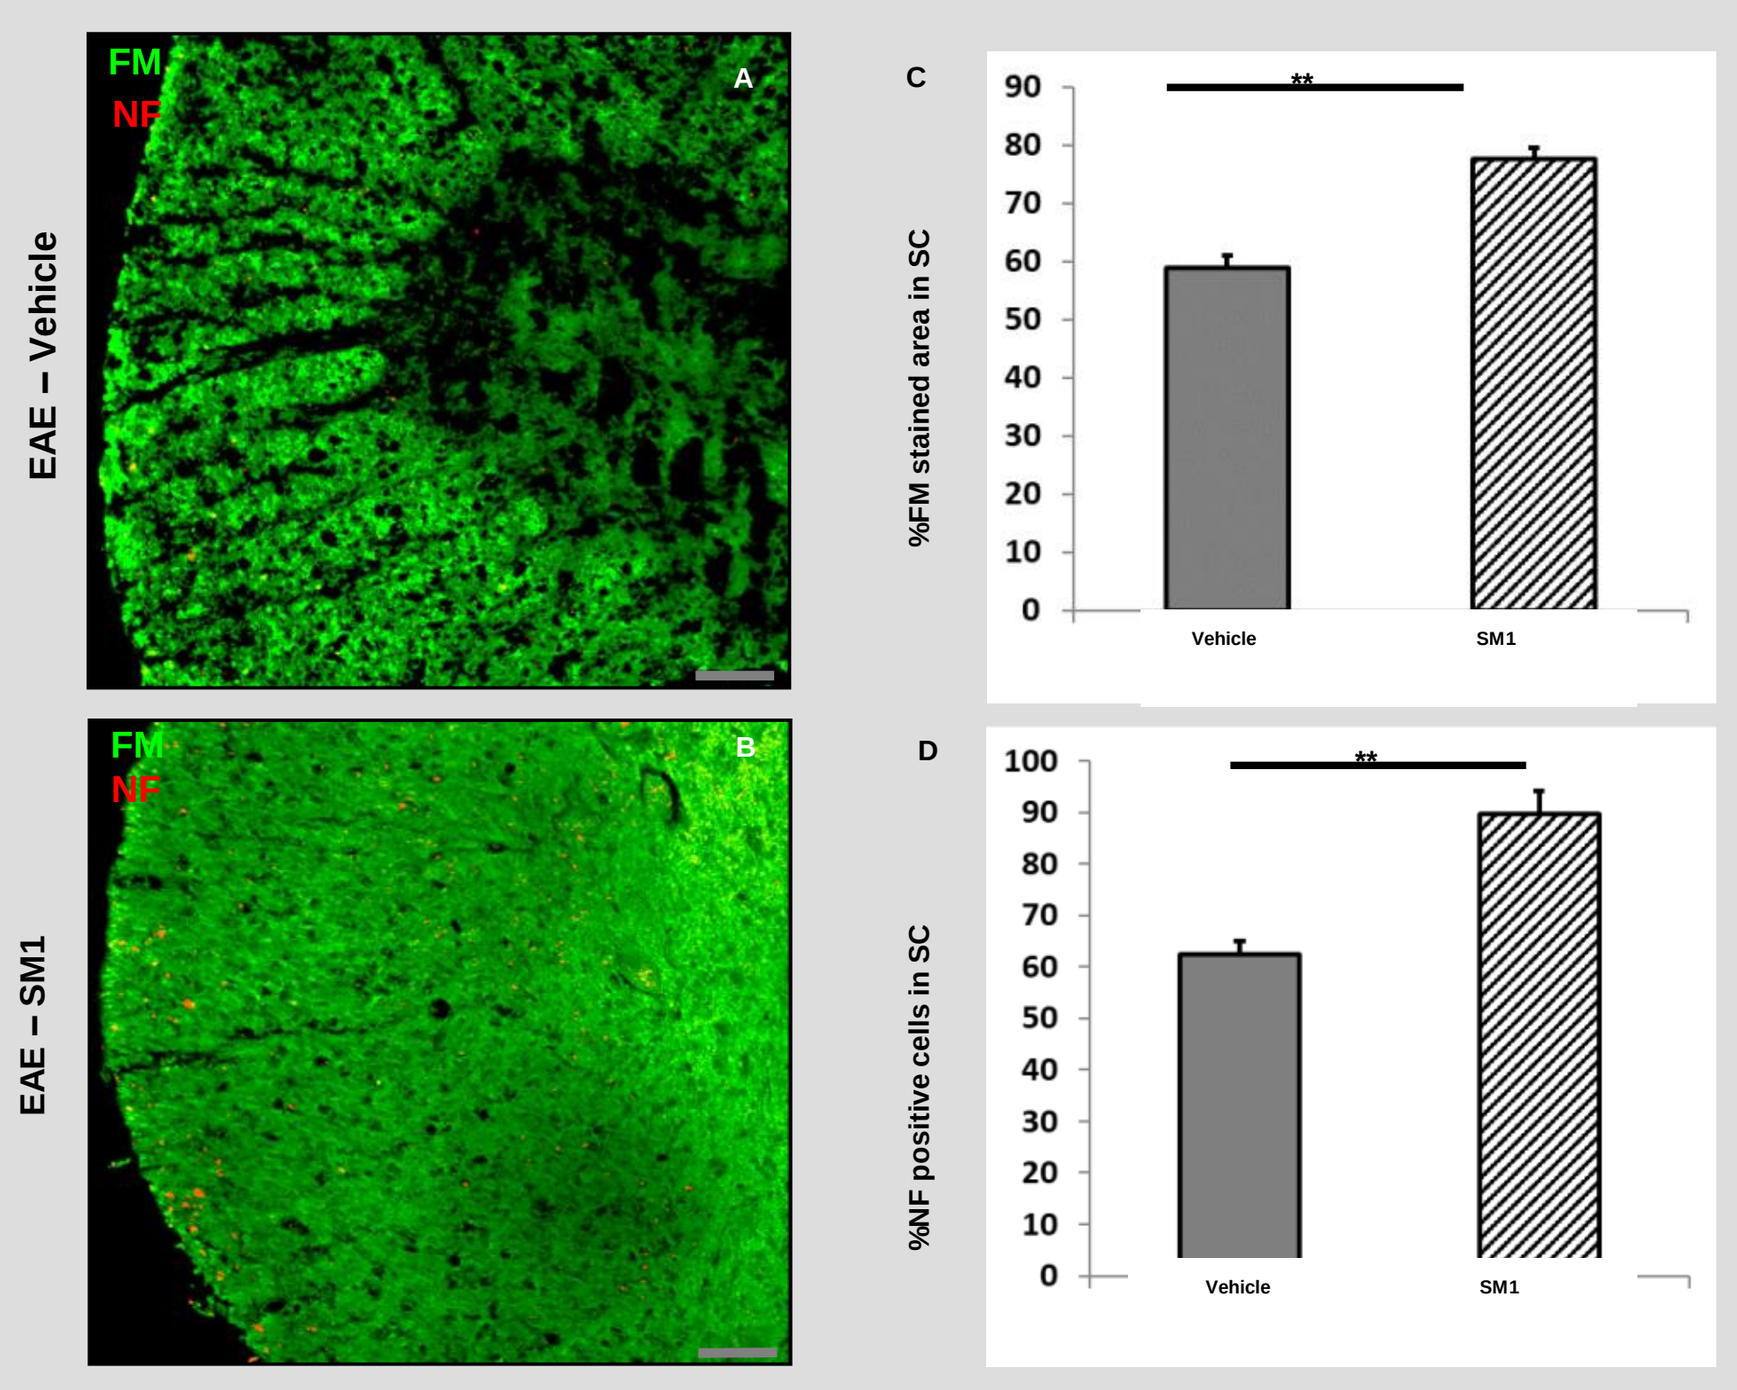
EAE – Vehicle
EAE – SM1
FM
NF
%FM stained area in SC
%NF positive cells in SC
Vehicle
SM1

Figure 2. ***SM administration to EAE mice increases myelination and axonal density.*** Oral treatment with SM significantly increase the percentage of myelinated cells (flouromyelin, FM) (B) and axonal density (neurofilament, NF) compared to vehicle treated mice. Percentage of positive cells per area is analyzed for FM (C) and NF (D). Scale bars= 50µm; * $p < 0.05$, ** $p < 0.01$.

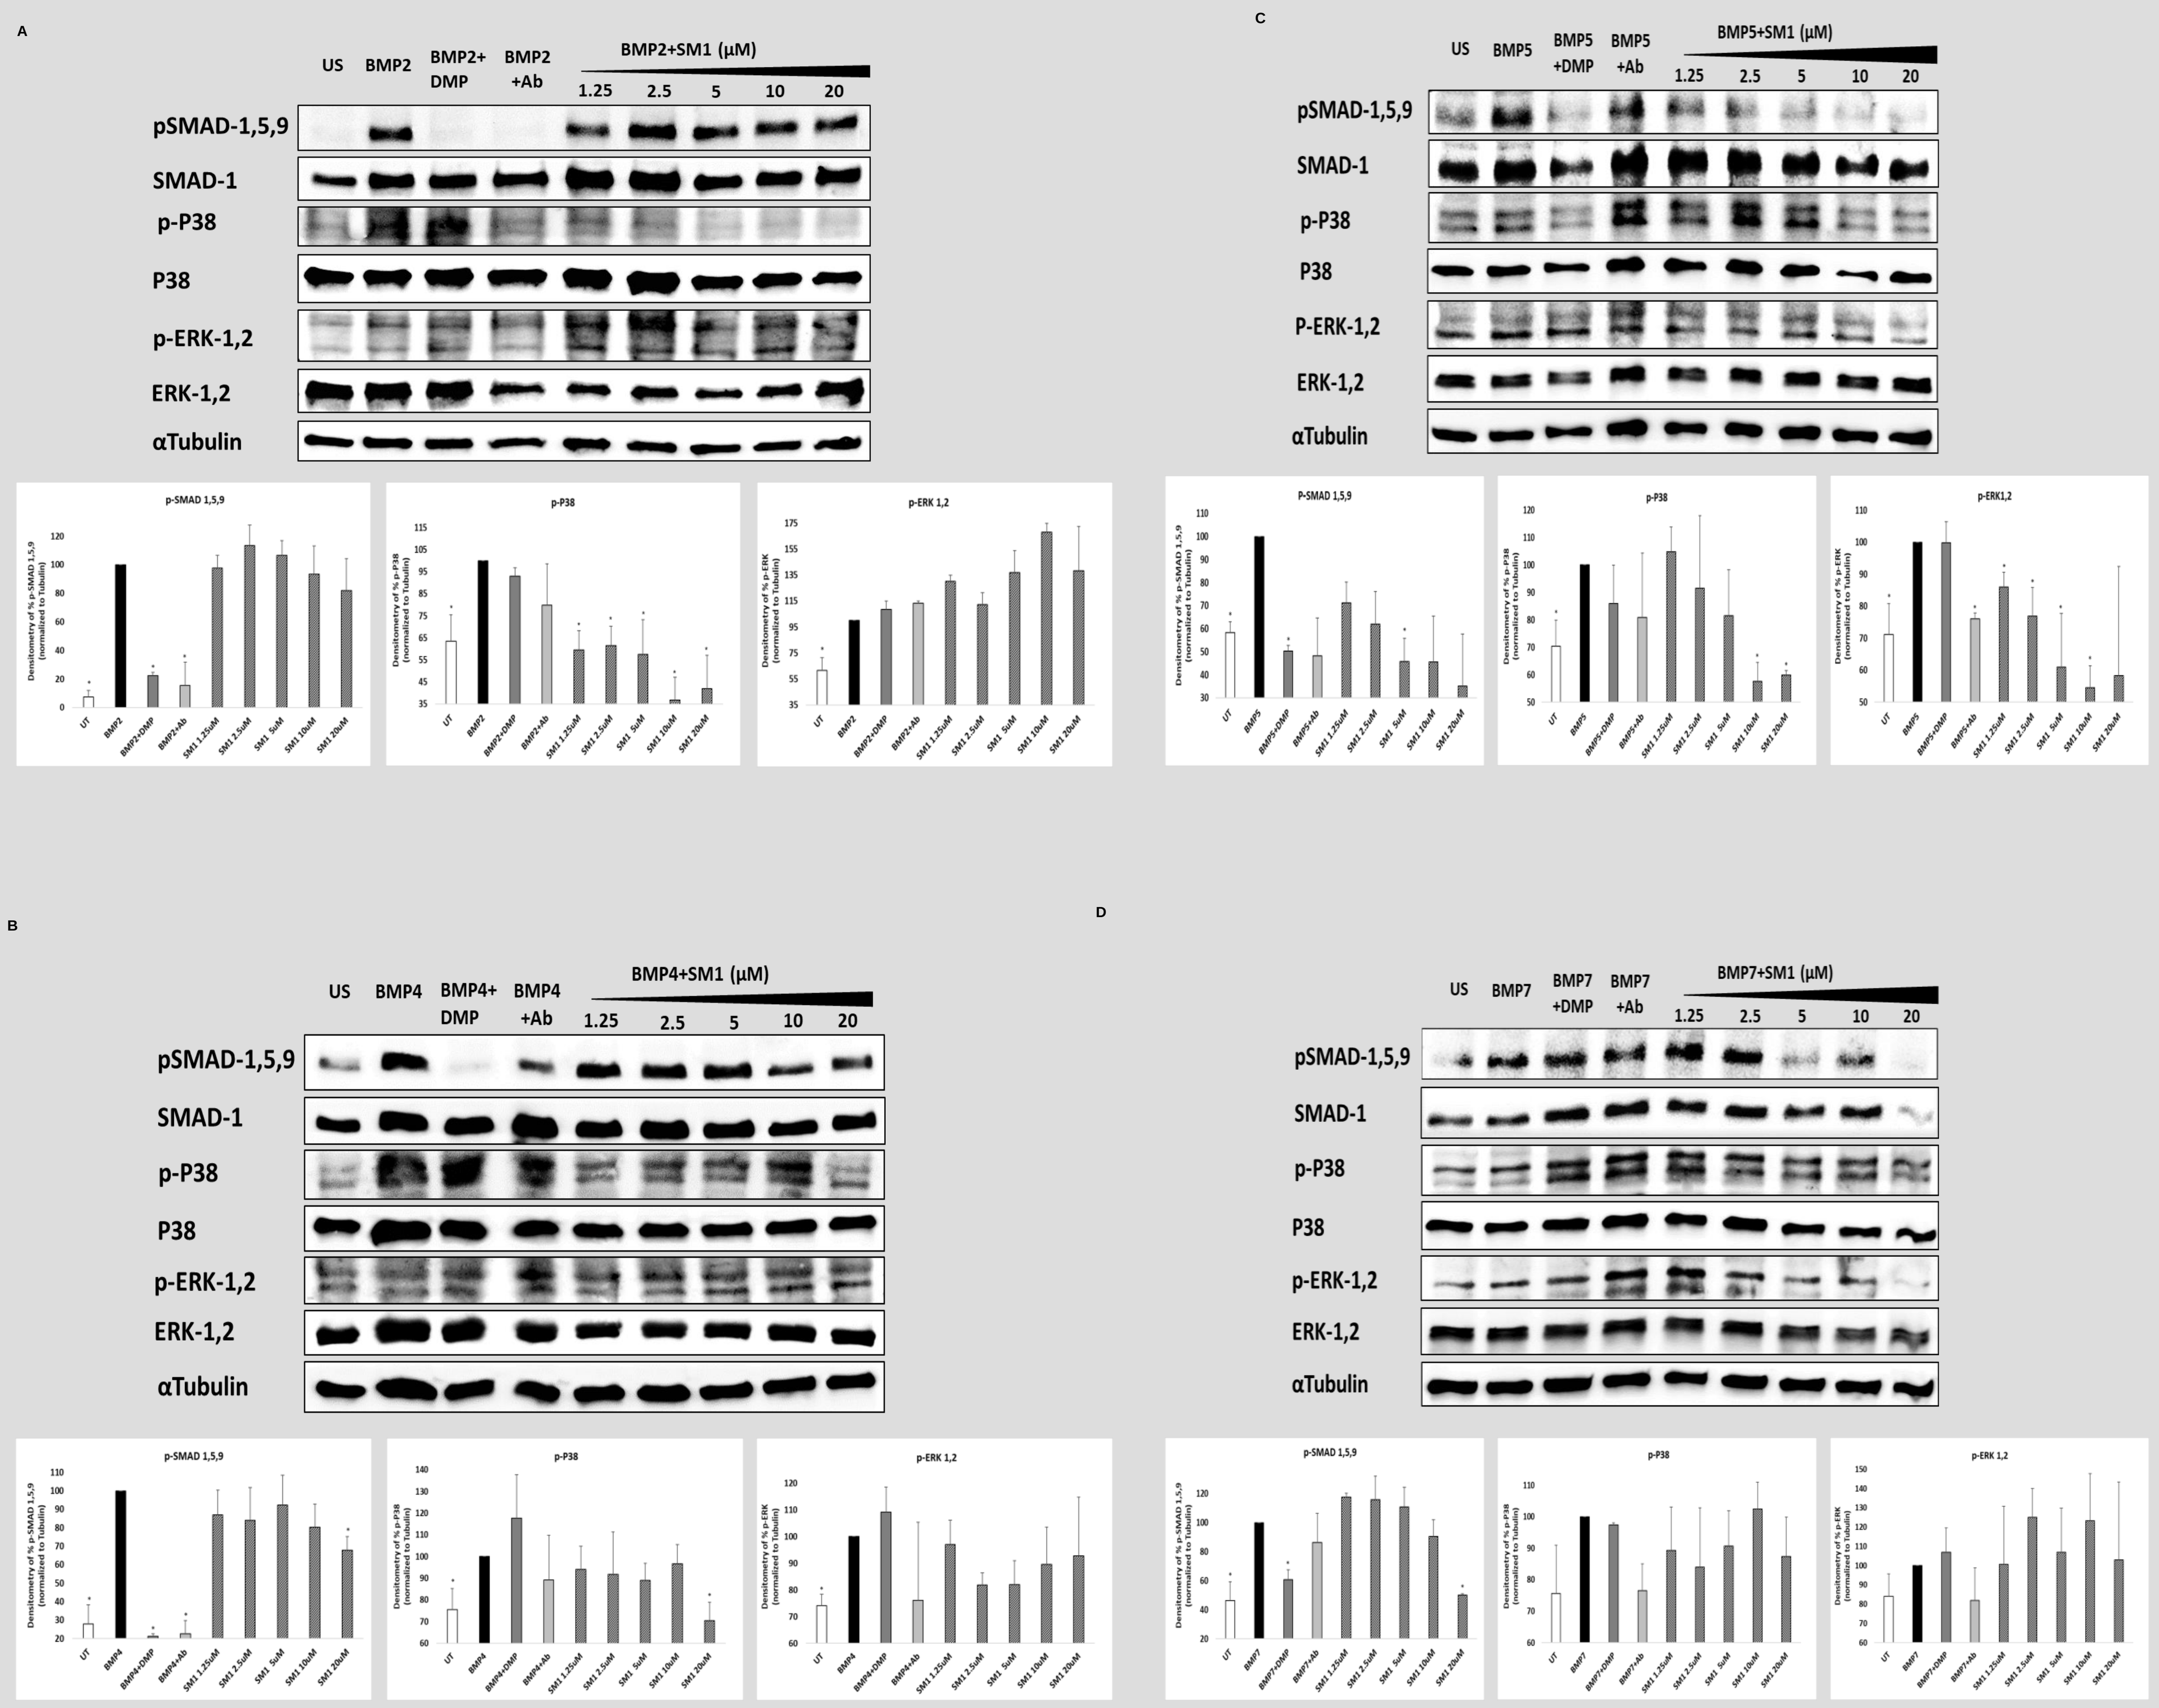
US
BMP2
BMP2+ DMP
BMP2 +Ab
BMP2+SM1 (µM)
BMP4+SM1 (µM)
BMP5+SM1 (µM)
BMP7+SM1 (µM)
1.25
2.5
5
10
20
pSMAD-1,5,9
SMAD-1
p-P38
P38
p-ERK-1,2
ERK-1,2
αTubulin

Figure 3. ***SM inhibits canonical and non-canonical BMP5 and non-canonical BMP2 signaling.*** Phosphorylation of SMAD1/5/9(8), MAPK p38 and ERK1,2 in NIH3T3 cells untreated or treated with BMP2 (A), BMP4 (B), BMP5 (C) or BMP7 (D) for 30 minutes, alone or pretreated with different concentrations of SM or 5µM dorsomorphin (DMP) or 500ng/ml blocking Ab as controls for 30 minutes. Western blot analysis was performed on the lysates and representative experiments are shown. The analysis was done on densitometry of 2-4 experiments, normalized to α-tubulin and BMPs treatment alone was set as 100%. * $p < 0.05$, ** $p < 0.01$.

CONCLUSIONS

- SM treatment promotes myelin and axonal density, while simultaneously increases the oligodendrogenesis and inhibits the astrogenesis in EAE.
- SM suppresses both canonical and non-canonical BMP5 and BMP2 signaling pathways in a BMP-specific manner.
- Therefore, SM may be an effective treatment for repairing lesions of demyelinating diseases.