Blockage of BMP-2/4 Signaling by Neutralizing Ab or Small Molecule Induces Remyelination and Oligodendrocyte Differentiation in Cuprizone Model of Demyelination

Moshe Benhamou[1,2], Karin Mausner-Fainberg[1], Maya Golan[1], Yona Gefen[3], Uri Danon[3], Ehud Marom[3], Arnon Karni[1,2,4]

[1]Neuroimmunology Laboratory, Department of Neurology, Tel Aviv Sourasky Medical Center; [2]Sackler's Faculty of Medicine, [3]Stem Cell Medicine Ltd. [4]Segol School of Neuroscience, Tel Aviv University, Israel.

BACKGROUND

- Oligodendrocyte regeneration and myelin repair are important goals in multiple sclerosis.
- Bone morphogenetic factors (BMPs) are factors that block oligodendrogenesis and remyelination.
- Increased BMP expression was observed in animal models of demyelination induced by cuprizone, ethidium bromide, or lysolecithin.

OBJECTIVE

To examine the effect of BMP-2 blockade, by anti-BMP-2/4 neutralizing Ab or by a BMP-2- inhibiting small molecule (SM) on remyelination and oligodendrocytes differentiation in the cuprizone model.

METHODS

- We used high throughput screening (HTS) for sorting of SMs that block BMP-2- induced-alkaline phosphatase (ALP) production in ATDC5 chondrogenic cells. SM1, from Maybridge library, was found to have an optimal IC50 (2.6µM), with an ameliorating effect in experimental autoimmune encephalomyelitis (EAE), similarly to the anti- BMP-2/4 neutralizing Ab.

SM1 discovery and its effect in relapsing EAE

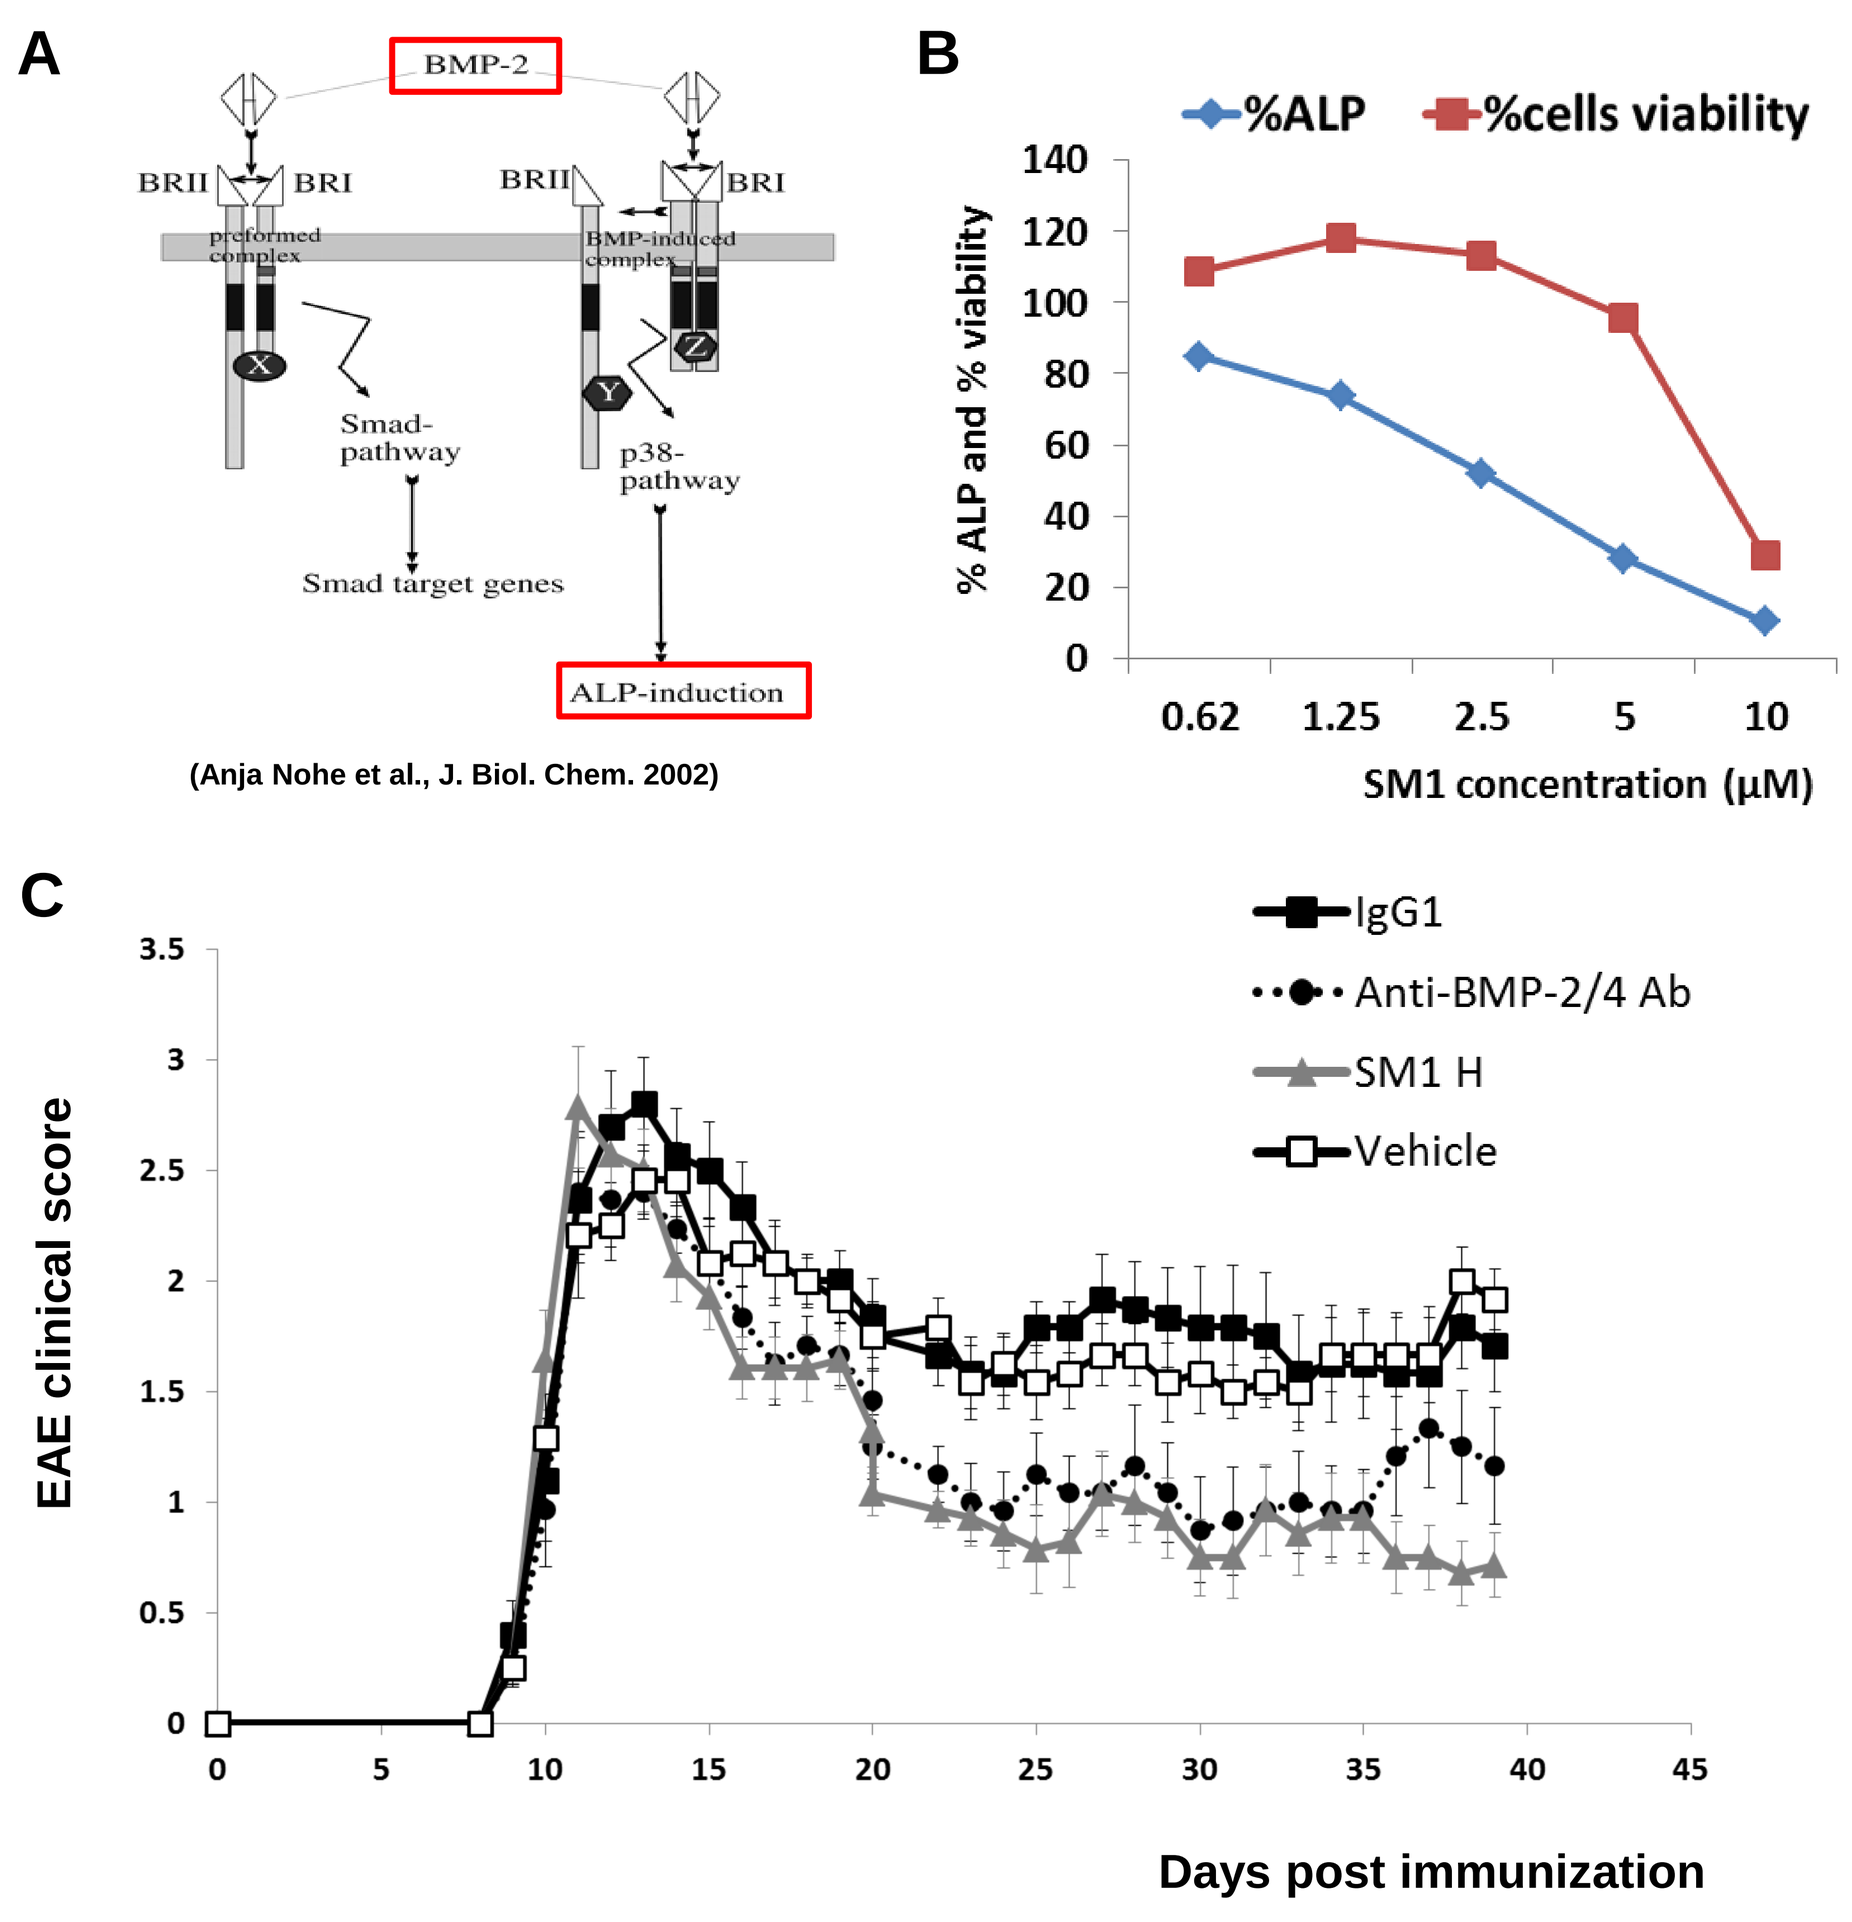

A
BMP-2
BRII
BRI
preformed complex
BMP-induced complex
Smad-pathway
p38-pathway
Smad target genes
ALP-induction
(Anja Nohe et al., J. Biol. Chem. 2002)
B
%ALP
%cells viability
% ALP and % viability
0.62
1.25
2.5
5
10
SM1 concentration (µM)
C
IgG1
Anti-BMP-2/4 Ab
SM1 H
Vehicle
EAE clinical score
Days post immunization

D

	Vehicle	SM1 H- 500 µg/mouse	IgG1	Anti-BMP-2/4 mAb 30µg/mouse
Incidence (score ≥ 1)	100%	92.8%	100.0%	100%
Disease onset (days)	9.5 ± 0.2	9.6 ± 0.1	9.5 ± 0.1	9.7 ± 0.2
Maximum score	2.8 ± 0.2	2.9 ± 0.3	3.2 ± 0.2	2.9 ± 0.2
Cumulative score	54.3 ± 2.6	37.9 ± 3.6	57.7 ± 4.3	41.1 ± 4.7
% amelioration in cumulative score	-	30.2% vs. vehicle P=0.001	-	28.8% vs. IgG1 P=0.02

Figure 1. (A) Sorting for SMs that block BMP-2 signaling was based on the principle of BMP-2 neutralization bioassay. rhBMP-2 is known to induce alkaline phosphatase (ALP) production in ATDC5 cells, chondrogenic cell line, *via* p38- pathway. (B) Following screening of 7600 Maybridge compounds, we received 96 potential hits, i.e. % inhibition of ALP production by BMP-2 stimulation was higher than 25% and toxicity level was lower than 20%. These hits were re-examined in a dose-dependent manner for IC50 determination. SM1 demonstrated the top IC50 value (2.6µM). (C) Relapsing EAE (R-EAE)- induced mice were daily IP administrated with either SM1 (500 µg/mouse) or vehicle alone (5% DMSO in PBS) starting from day 9 post immunization for 30 following days. R-EAE mice were also IV administrated with anti- BMP-2/4 neutralizing mAb (R&D systems) or the corresponding IC, IgG1, on day 9 post-immunization (30 µg/mouse). The table presented in (D) summarizes the clinical characteristics of R-EAE mice. N=14 in each group.

RESULTS

I. Anti- BMP-2/4 mAb and SM1 promote remyelination in the corpus callosum of cuprizone- induced mice

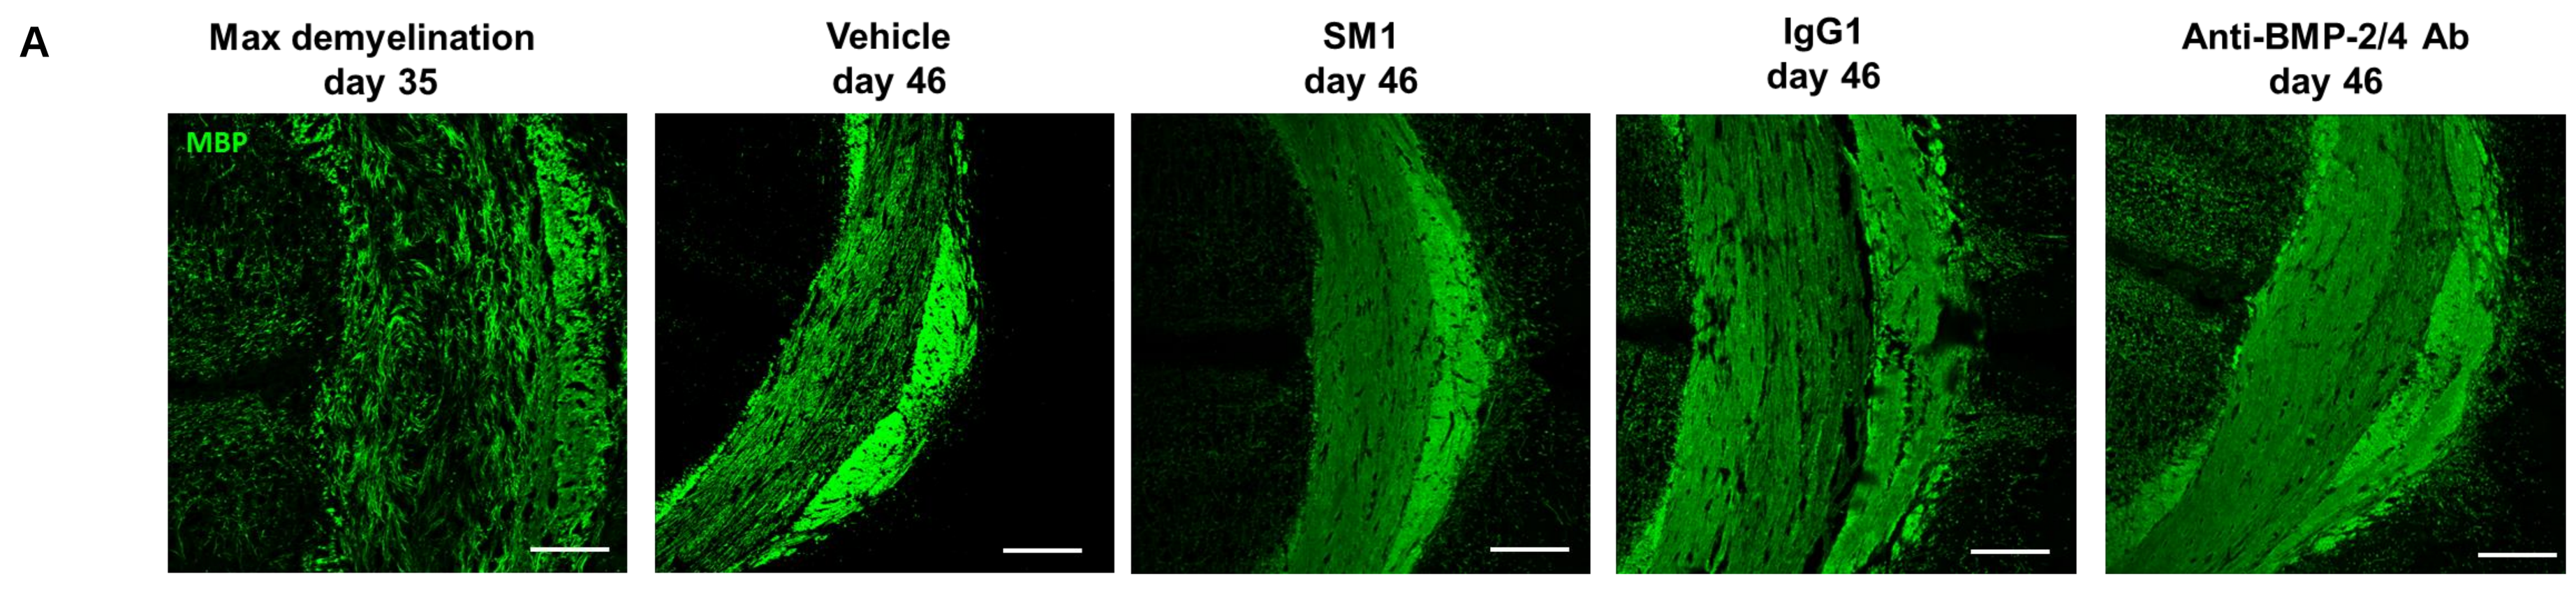

A
Max demyelination day 35
Vehicle day 46
SM1 day 46
IgG1 day 46
Anti-BMP-2/4 Ab day 46
MBP

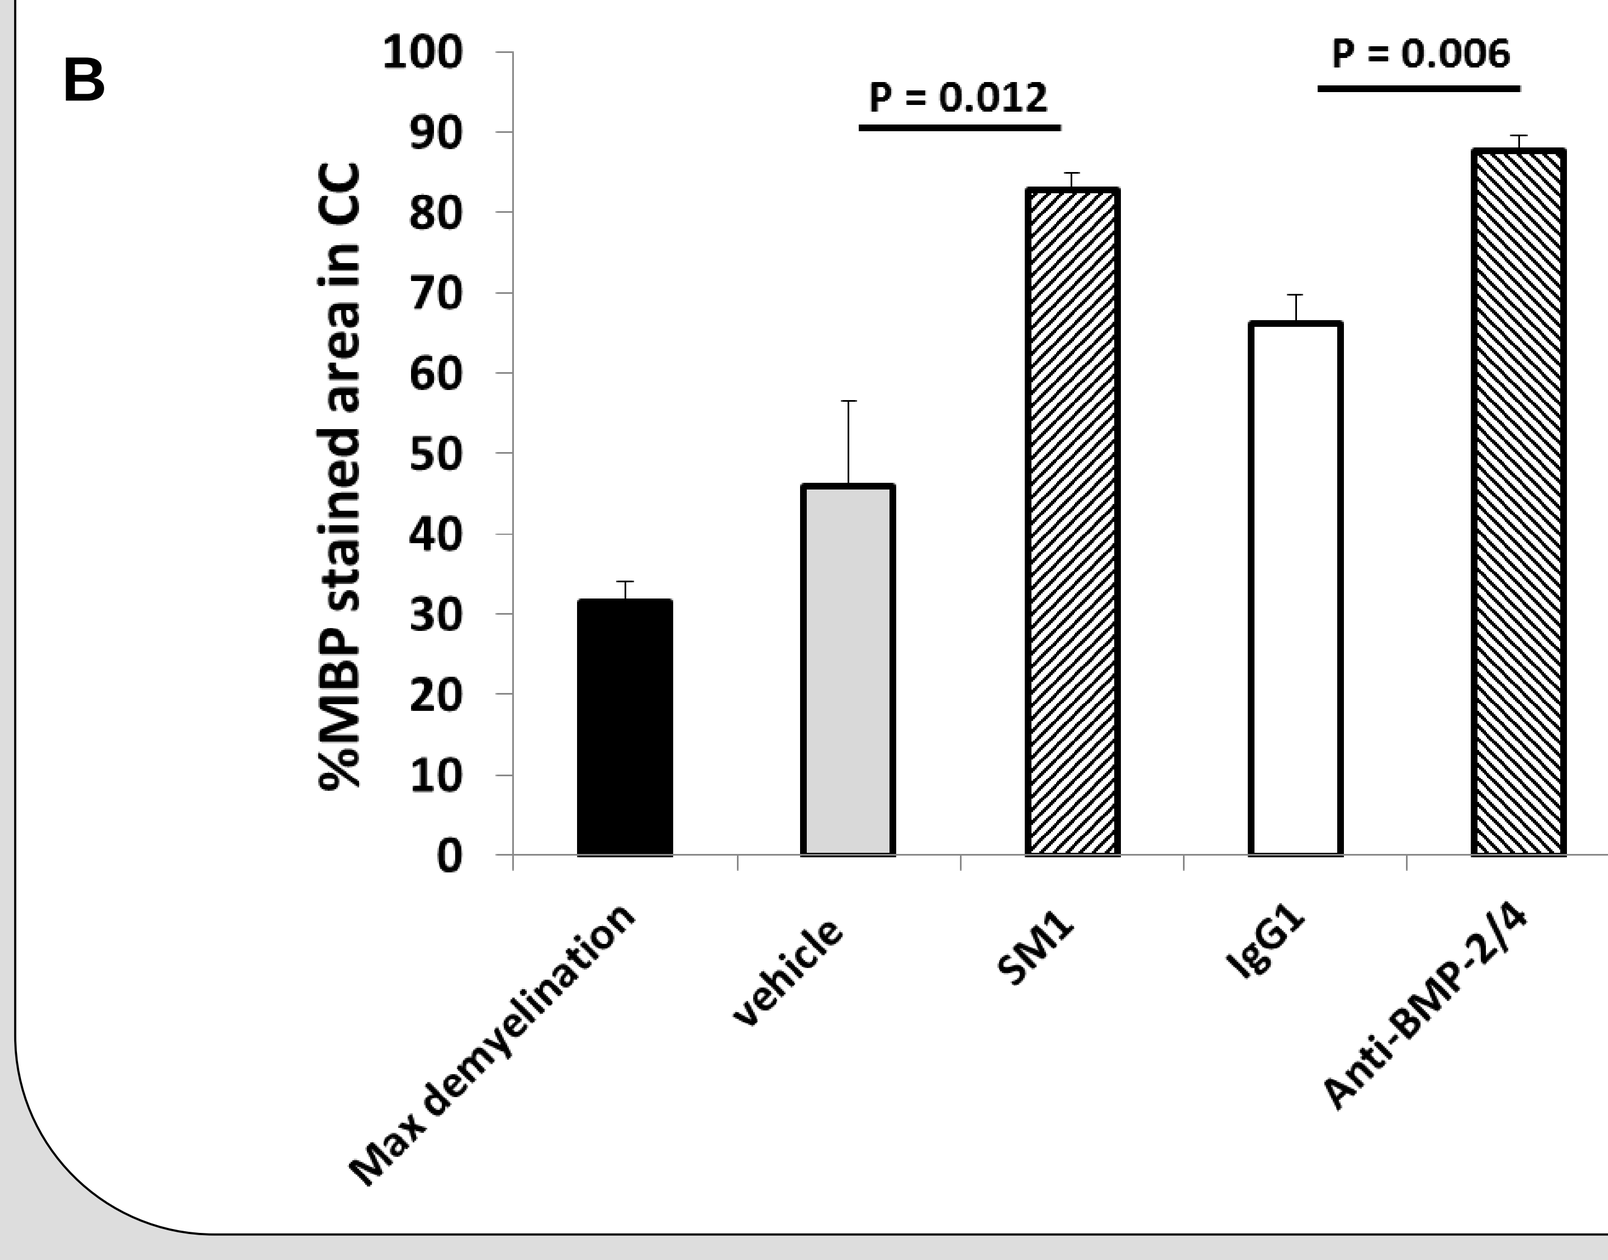

B
%MBP stained area in CC
P = 0.012
P = 0.006
Max demyelination
vehicle
SM1
IgG1
Anti-BMP-2/4

Figure 2. (A) C57BL/6 mice were fed 0.2% cuprizone for 5 weeks (day 35, maximum demyelination) and then transferred to a regular diet. The anti-BMP-2/4 neutralizing mAb was IV administrated (30 µg/mouse) on day 15 after the initiation of the cuprizone diet. A control group was IV injected with 30 µg/mouse of mouse IgG1. SM1 (500 µg/mouse) or vehicle alone (5% DMSO in PBS) was daily IP administrated from day 15 for the 30 following days. Mice were sacrificed 10 days after resuming the regular diet (recovery phase, day 46), N = 5 in each group. Myelin basic protein (MBP) levels were examined in the corpus callosum (CC) by immunofluorescence, (scale bar = 100µm).
(B) Quantification of % stained area was performed using *imageJ* software.

II. Anti- BMP-2/4 mAb and SM1 enhance OPCs differentiation into mature Olig-2⁺CC1⁺ oligodendrocytes in the corpus callosum of cuprizone- induced mice

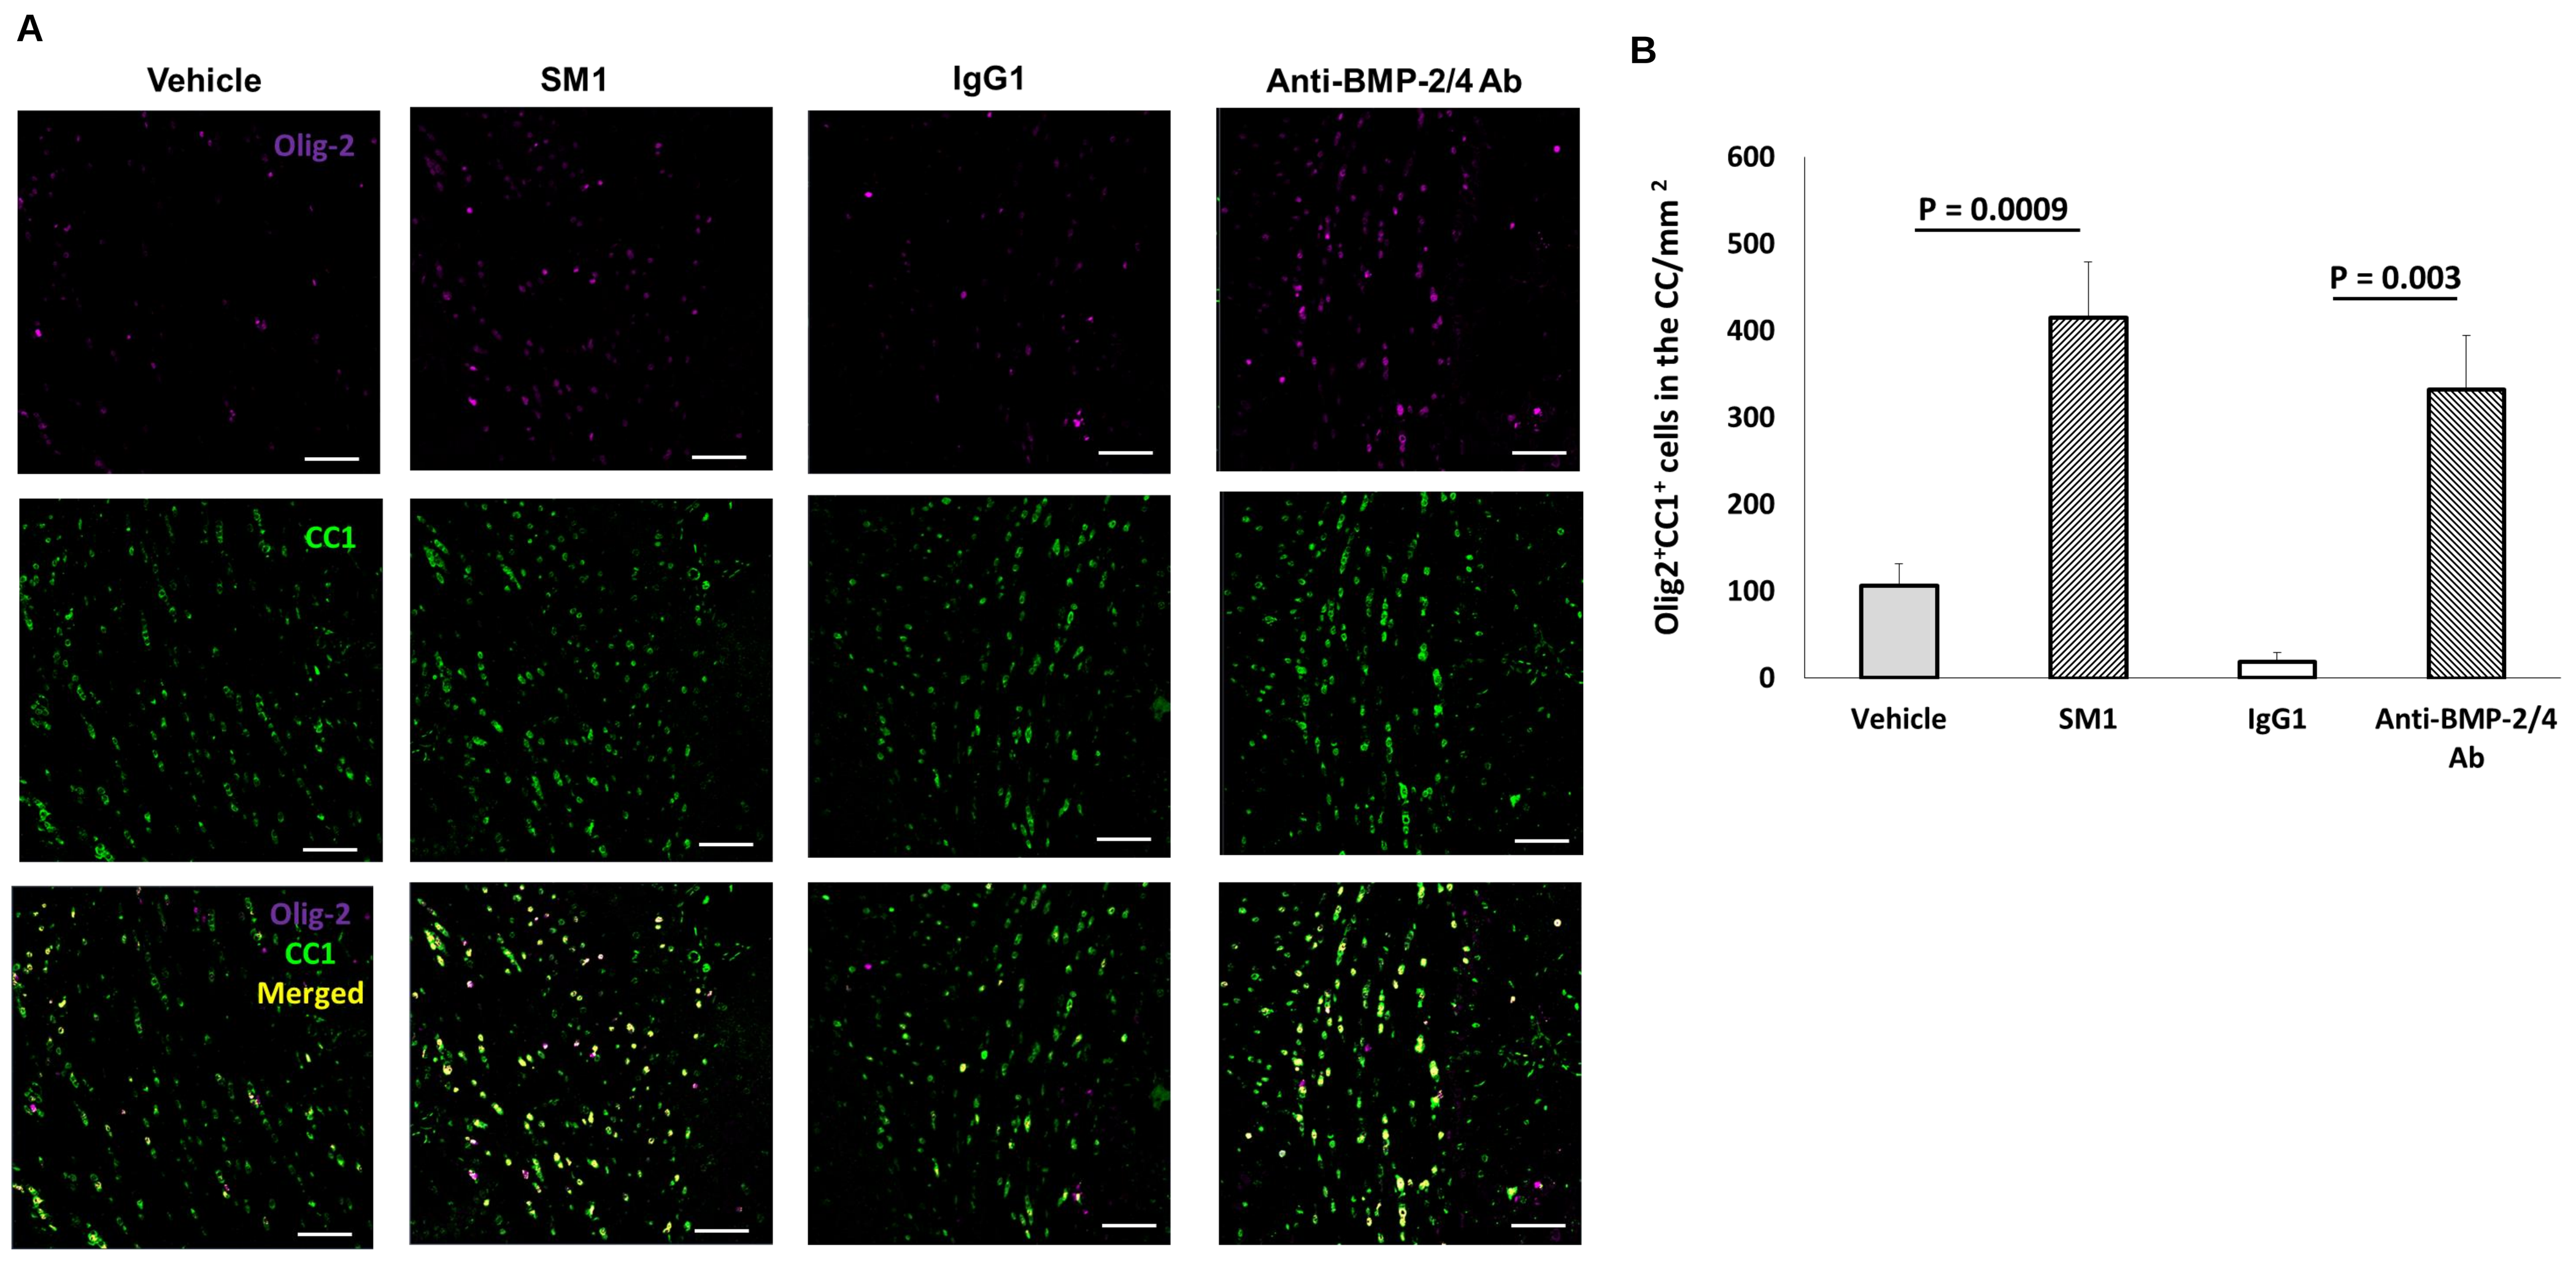

A
Vehicle
SM1
IgG1
Anti-BMP-2/4 Ab
Olig-2
CC1
Olig-2
CC1
Merged
B
Olig2⁺CC1⁺ cells in the CC/mm²
P = 0.0009
P = 0.003
Vehicle
SM1
IgG1
Anti-BMP-2/4 Ab

Figure 3. (A) Double- stained Olig-2+CC1+ cells were examined on day 46 in the corpus callosum (CC) of cuprizone- induced mice that were IP treated with either vehicle (5% DMSO in PBS) or SM1 (500 µg/mouse), from day 15 post cuprizone initiation for 30 following days, or that were IV treated with either anti- BMP-2/4 neutralizing mAb (30 µg/mouse) or isotype control (IgG1), on day 15 post cuprizone initiation (scale bar = 50µm). (B) The number of double-positive cells was quantified and was calculated as the number of cells/mm^2.

III. Anti- BMP-2/4 mAb and SM1 reduce SMAD phosphorylation in the corpus callosum of cuprizone- induced mice

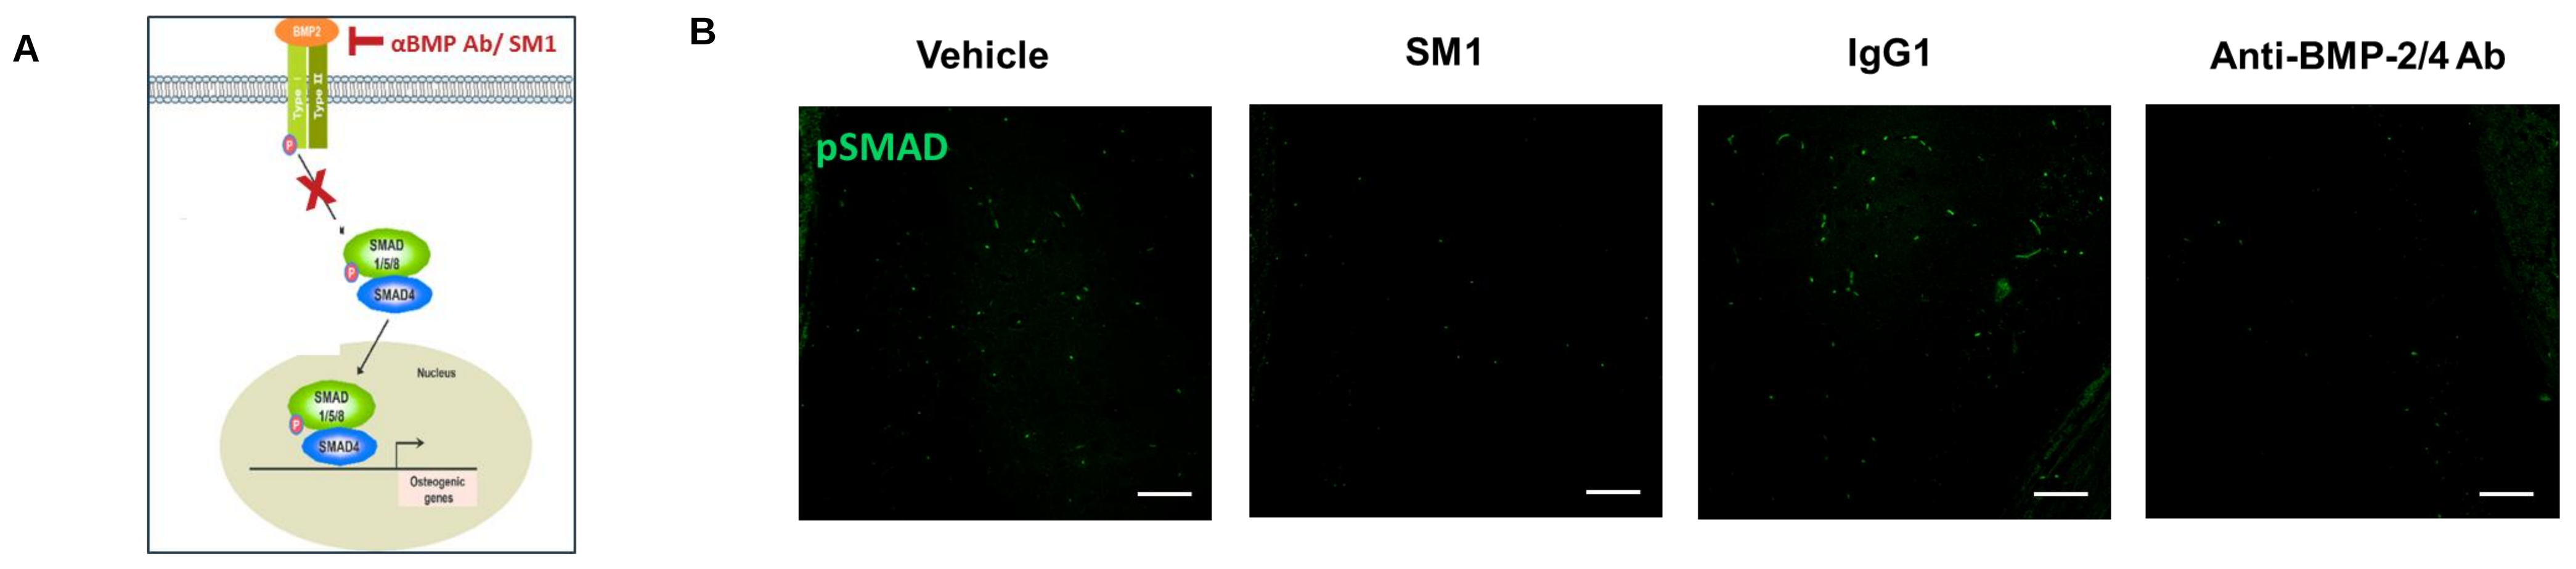

A
BMP2
αBMP Ab/ SM1
Type I
Type II
SMAD 1/5/8
SMAD4
Nucleus
Osteogenic genes
B
Vehicle
SM1
IgG1
Anti-BMP-2/4 Ab
pSMAD

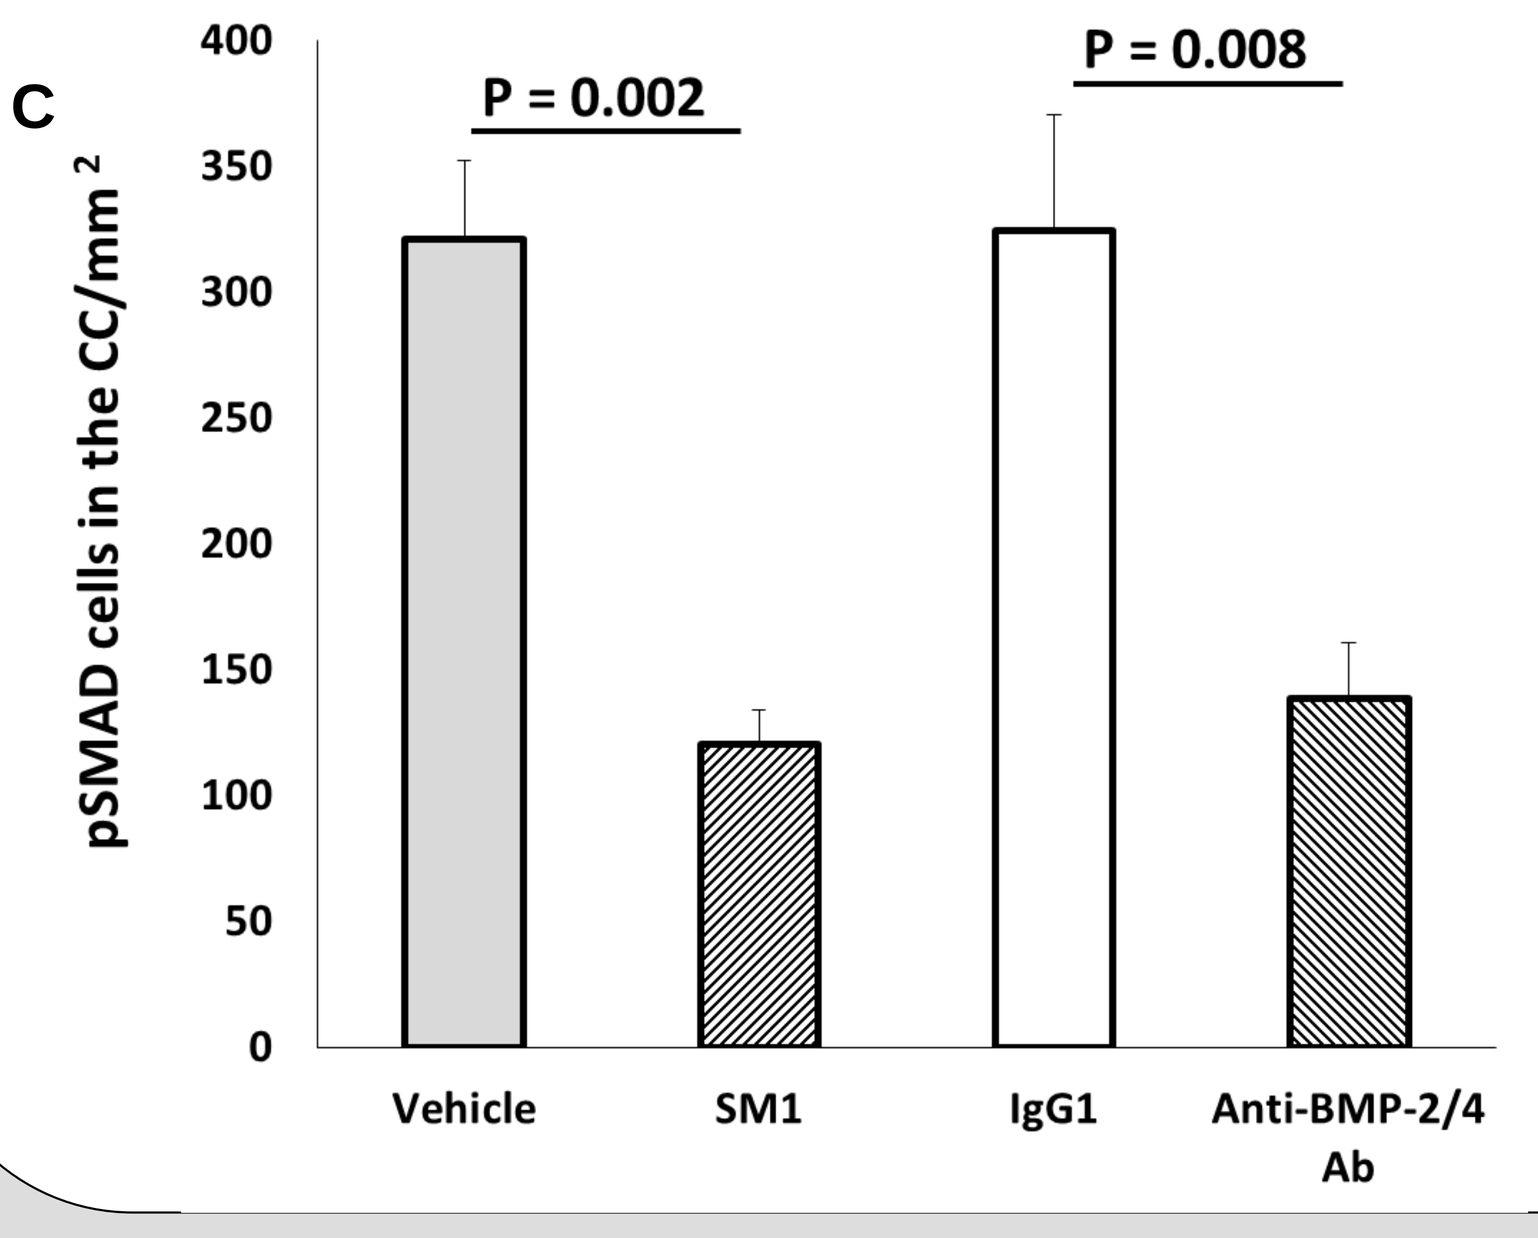

C
pSMAD cells in the CC/mm²
P = 0.002
P = 0.008
Vehicle
SM1
IgG1
Anti-BMP-2/4 Ab

Figure 4. (A) Schematic illustration of the canonical SMAD signaling in response to BMP-2 stimulation. (B) Phosphorylated SMAD (pSMAD+) cells were examined on day 46 in the corpus callosum (CC) of cuprizone- induced mice that were IP treated with either vehicle (5% DMSO in PBS) or SM1 (500 µg/mouse), for 30 following days, from day 15, as well as in cuprizone- induced mice that were IV treated with either anti- BMP-2/4 neutralizing mAb (30 µg/mouse) or IC- treated mice (IgG1), on day 15 post cuprizone initiation (scale bar = 50µm). (C) The number of pSMAD+ cells was quantified and was calculated as the number of cells/mm^2.

CONCLUSIONS

- **Systemic administration of either SM1 or anti- BMP-2/4 neutralizing Ab to cuprizone- induced mice induce enhanced myelination and oligodendrocytes maturation together with BMP2/4 signaling inhibition.**
- **Anti- BMP-2/4 Ab or SM1 are potential therapies for oligodendrocytes differentiation and remyelination in demyelinating disease such as multiple sclerosis.**

DISCLOSURE

The study was supported by Stem Cell Medicine Ltd.